Application of various types of linkers for photoaffinity biotin-tagged chemical probes to identify drug targets

M. Yuri[1], M. Hiramoto[1], S. Ujihara[1], N. Nakamura[1], T. Yamauchi[1], M. Naitou[1], M. Mabuchi[2,3], A. Tanaka[2,3], H. Yokota[1]

1: Drug Discovery Research, Astellas Pharma Inc, Japan
2: Advanced Medical Research Center, Hyogo University of Health Science, Japan
3: Department of Pharmacy, Hyogo University of Health Science, Japan

astellas
Leading Light for Life

Introduction

Target identification of drugs discovered in cell-based assays becomes very important in drug discovery, which is often performed by a chemical proteomics approach.

Recently, a tri-functional photoaffinity probe composed of a compound-binding part, a photoreactive part and a biotin-tagged part has been used for that purpose (1). In this approach, it is critical that the linker structure between the compound-binding and the photoreactive parts should not interfere with the binding between the compound and its target protein.

To examine the effect of the linker structure, we prepared tri-functional probes connected with a tool compound BisIII (2) with various type of linkers and we compared the quantities of the captured proteins by label-free quantitative proteomic analysis.

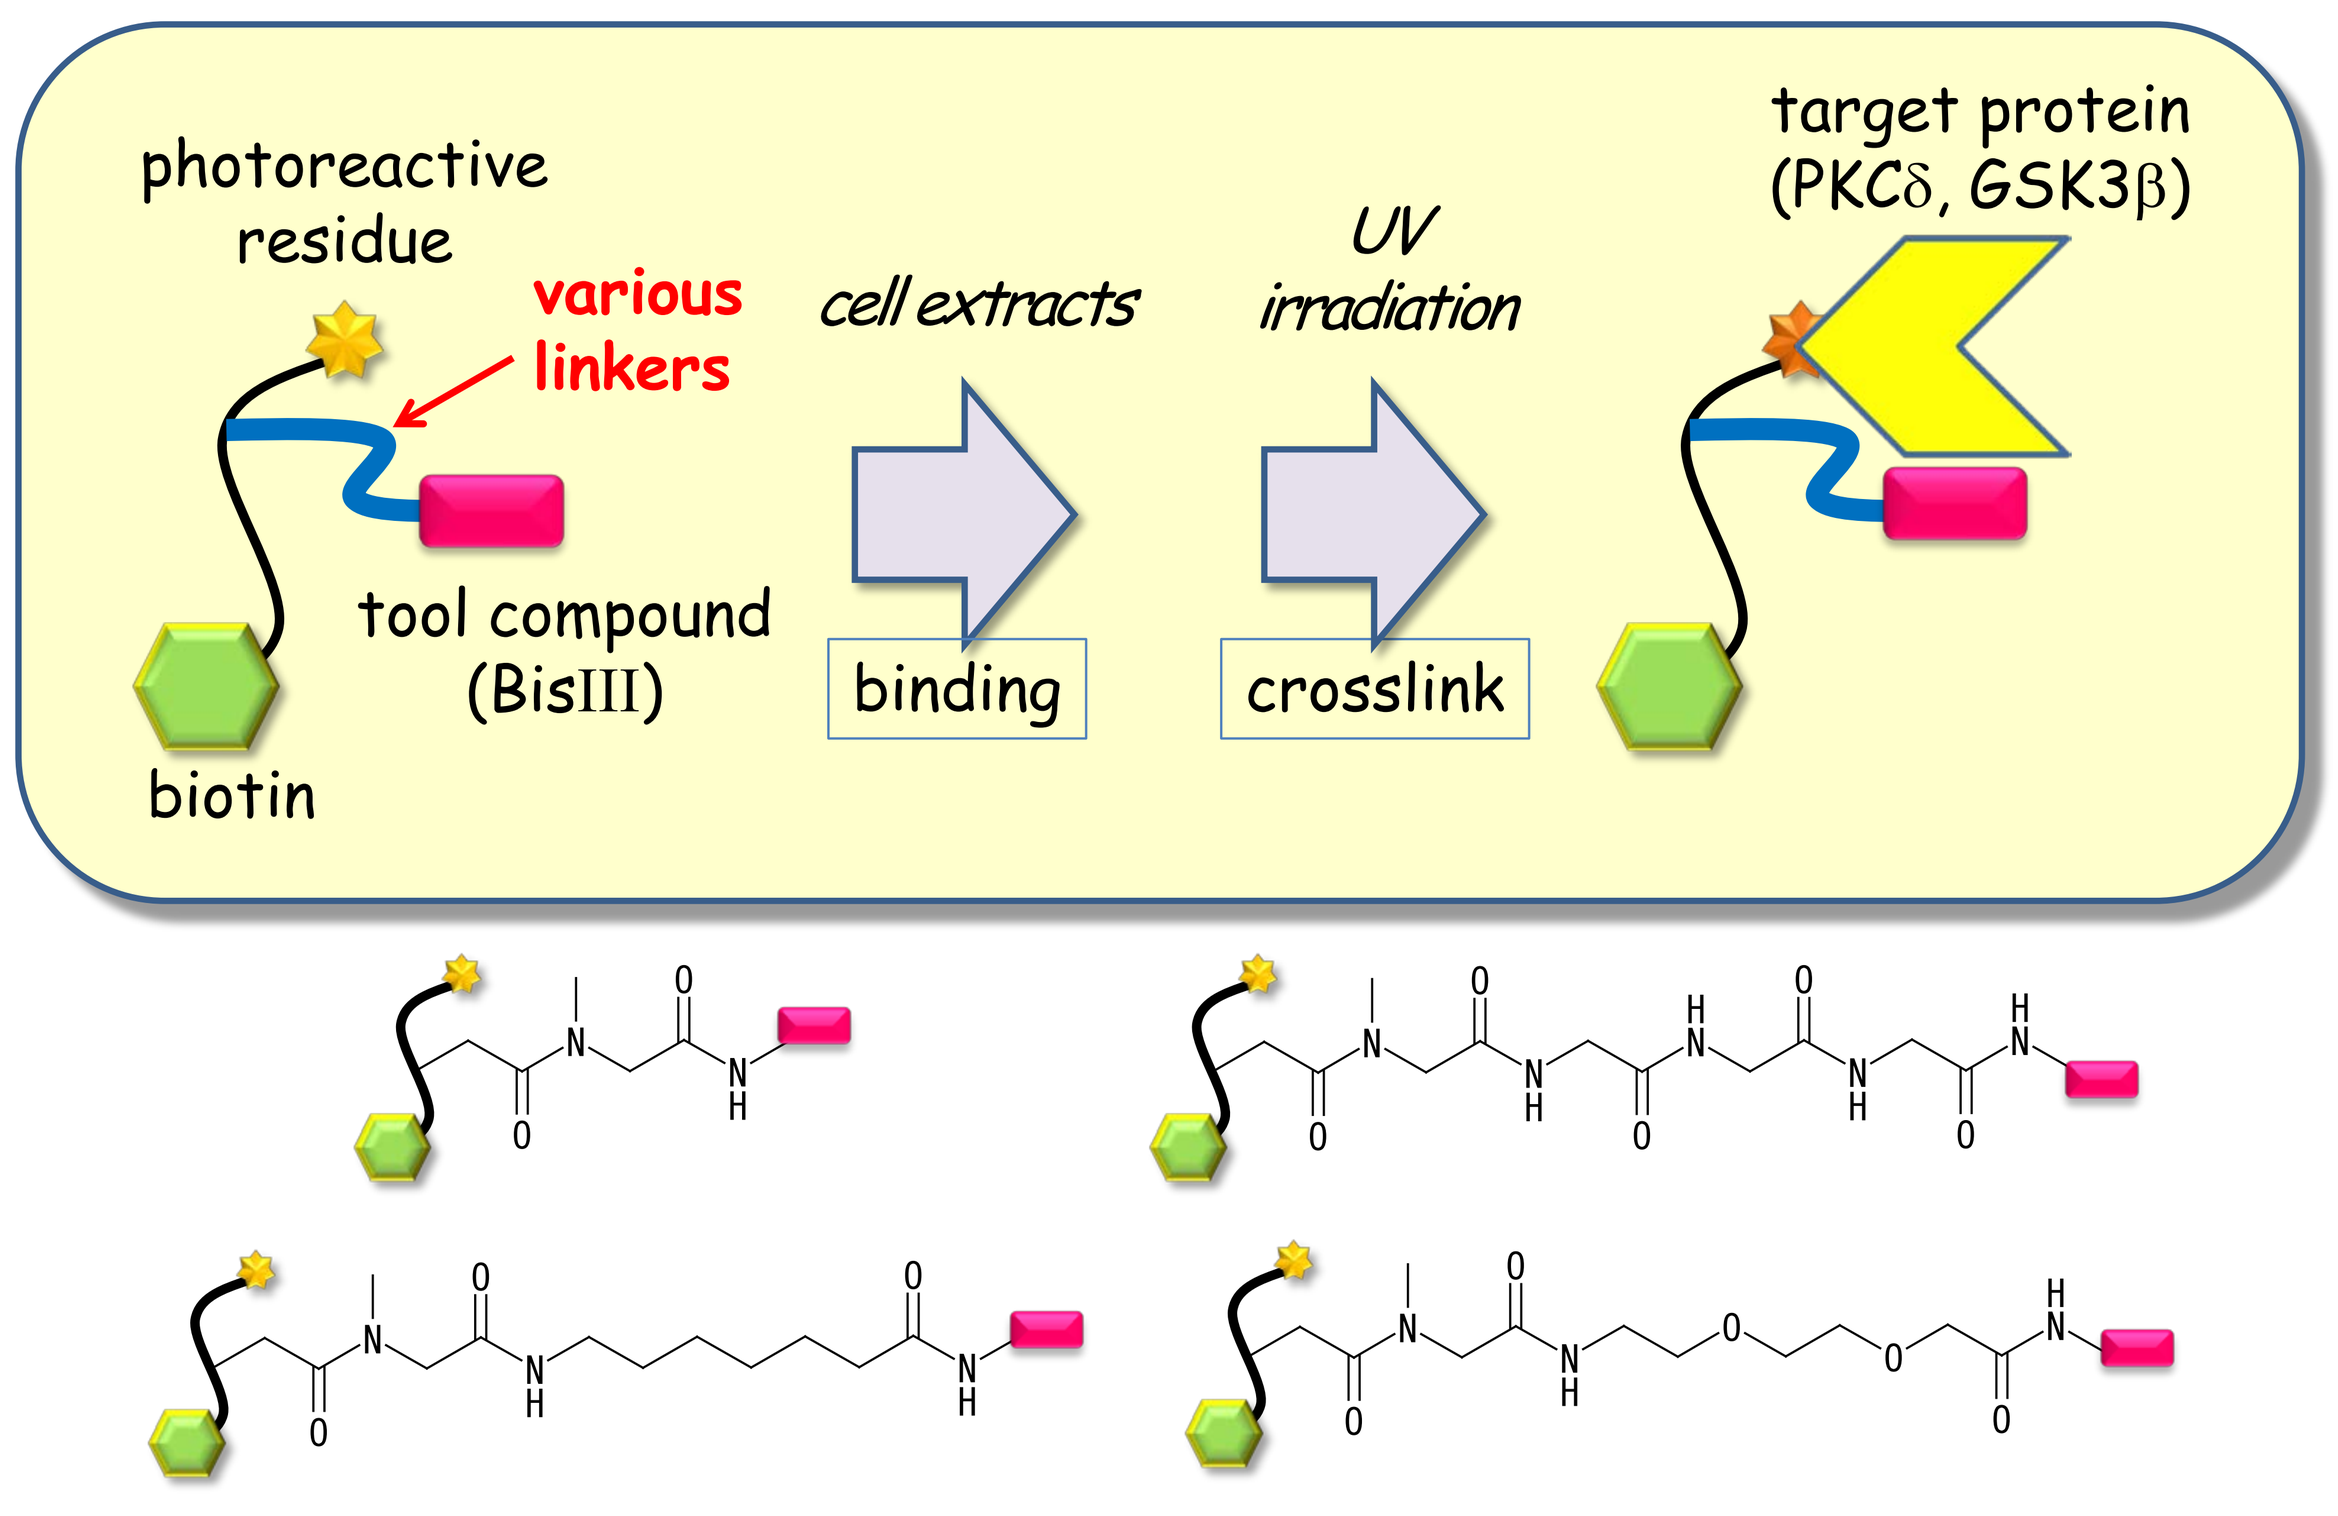
photoreactive residue
various linkers
tool compound (BisIII)
biotin
cell extracts
binding
UV irradiation
crosslink
target protein (PKCδ, GSK3β)

What are the captured proteins changed by using the probes which have a various kind of linkers?

Summary

We tested the tri-functional probes with a various kind of linkers. We found that the linker structure influenced the degree of the quantity of captured proteins, but the effects were different in the individual protein both of the target and non-specific proteins.

No one knows which linker is the best for the particular compounds which targets are unknown, so we think that it would be helpful for efficient target identification to use a combination of tri-functional probes with various kinds of linkers.

References

(1) J. Fisher et al, *J. Proteome Res.*, **2010**, *9*, 806-817.
(2) D. Brehmer et al. *Mol. Cell. Proteomics*, **2004**, *3*, 490-500

Results

1. Structure of the probe compounds

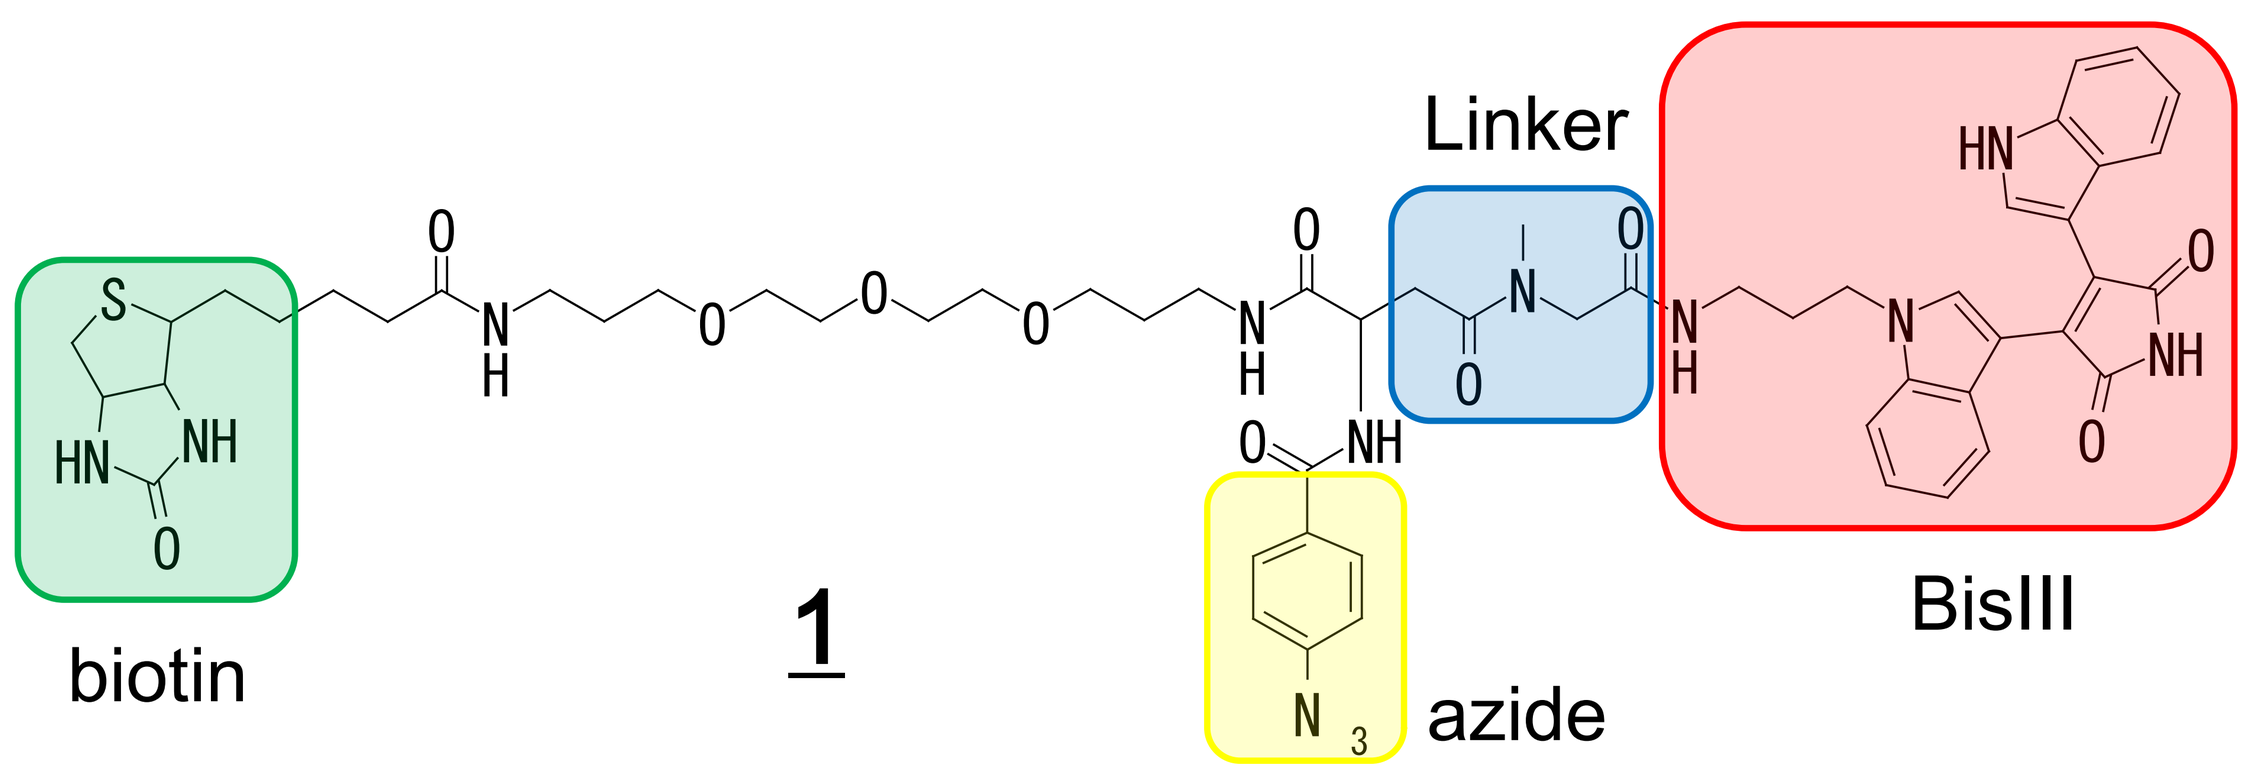
Linker
biotin
azide
BisIII

1

structures of the linker part

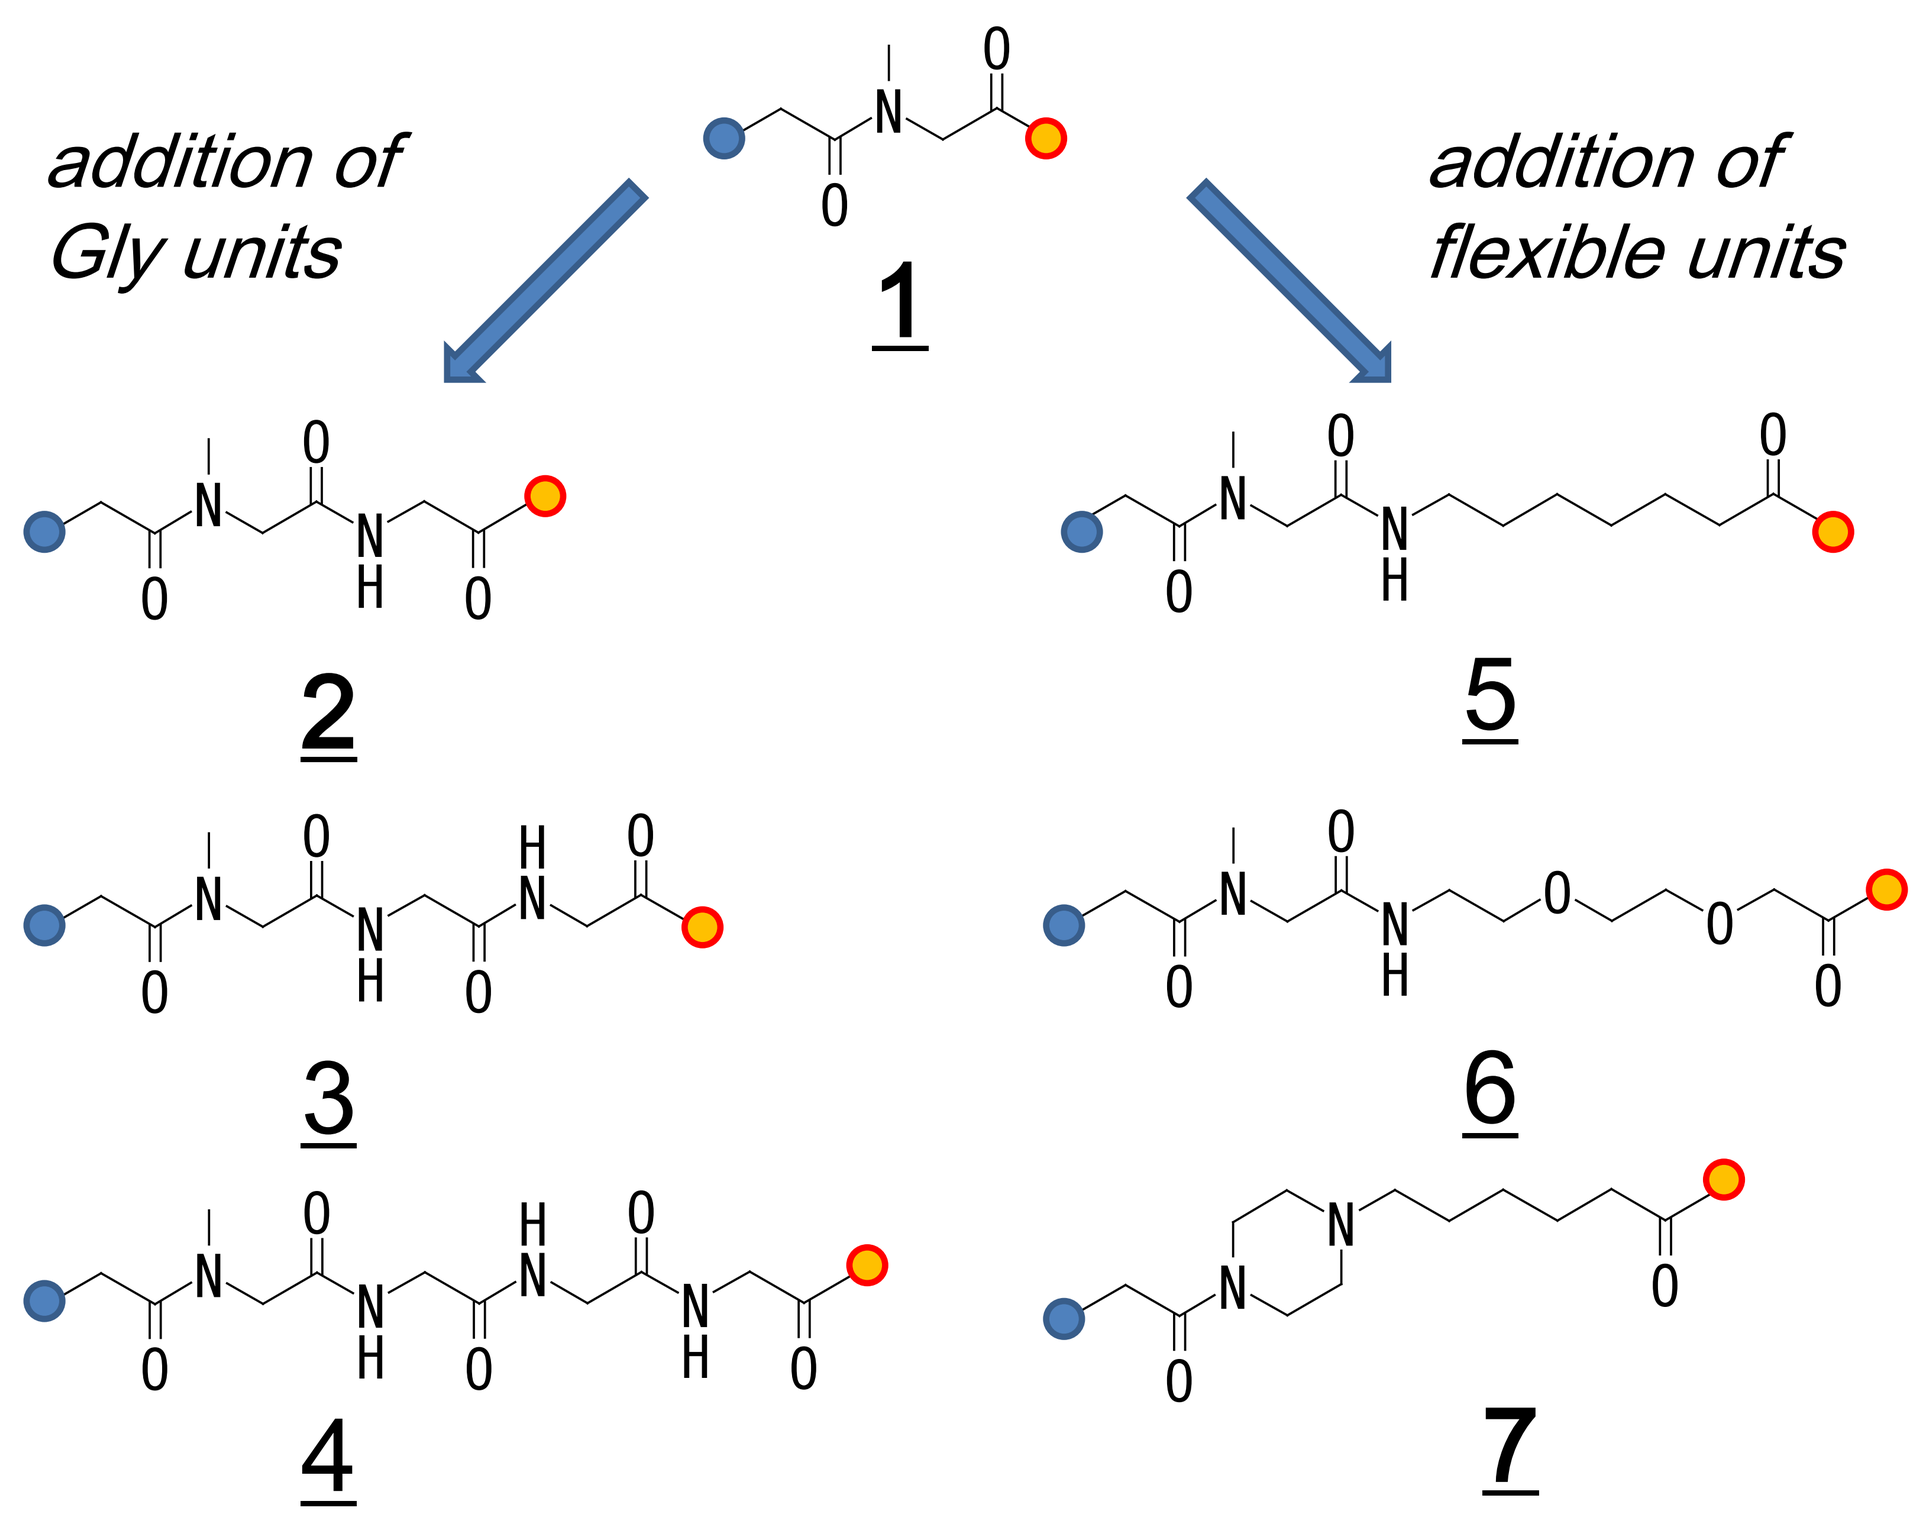
addition of Gly units
addition of flexible units
1
2
3
4
5
6
7

2. BisIII target proteins were captured using the probe1 with UV irradiation.

effects of UV irradiation

Western blotting

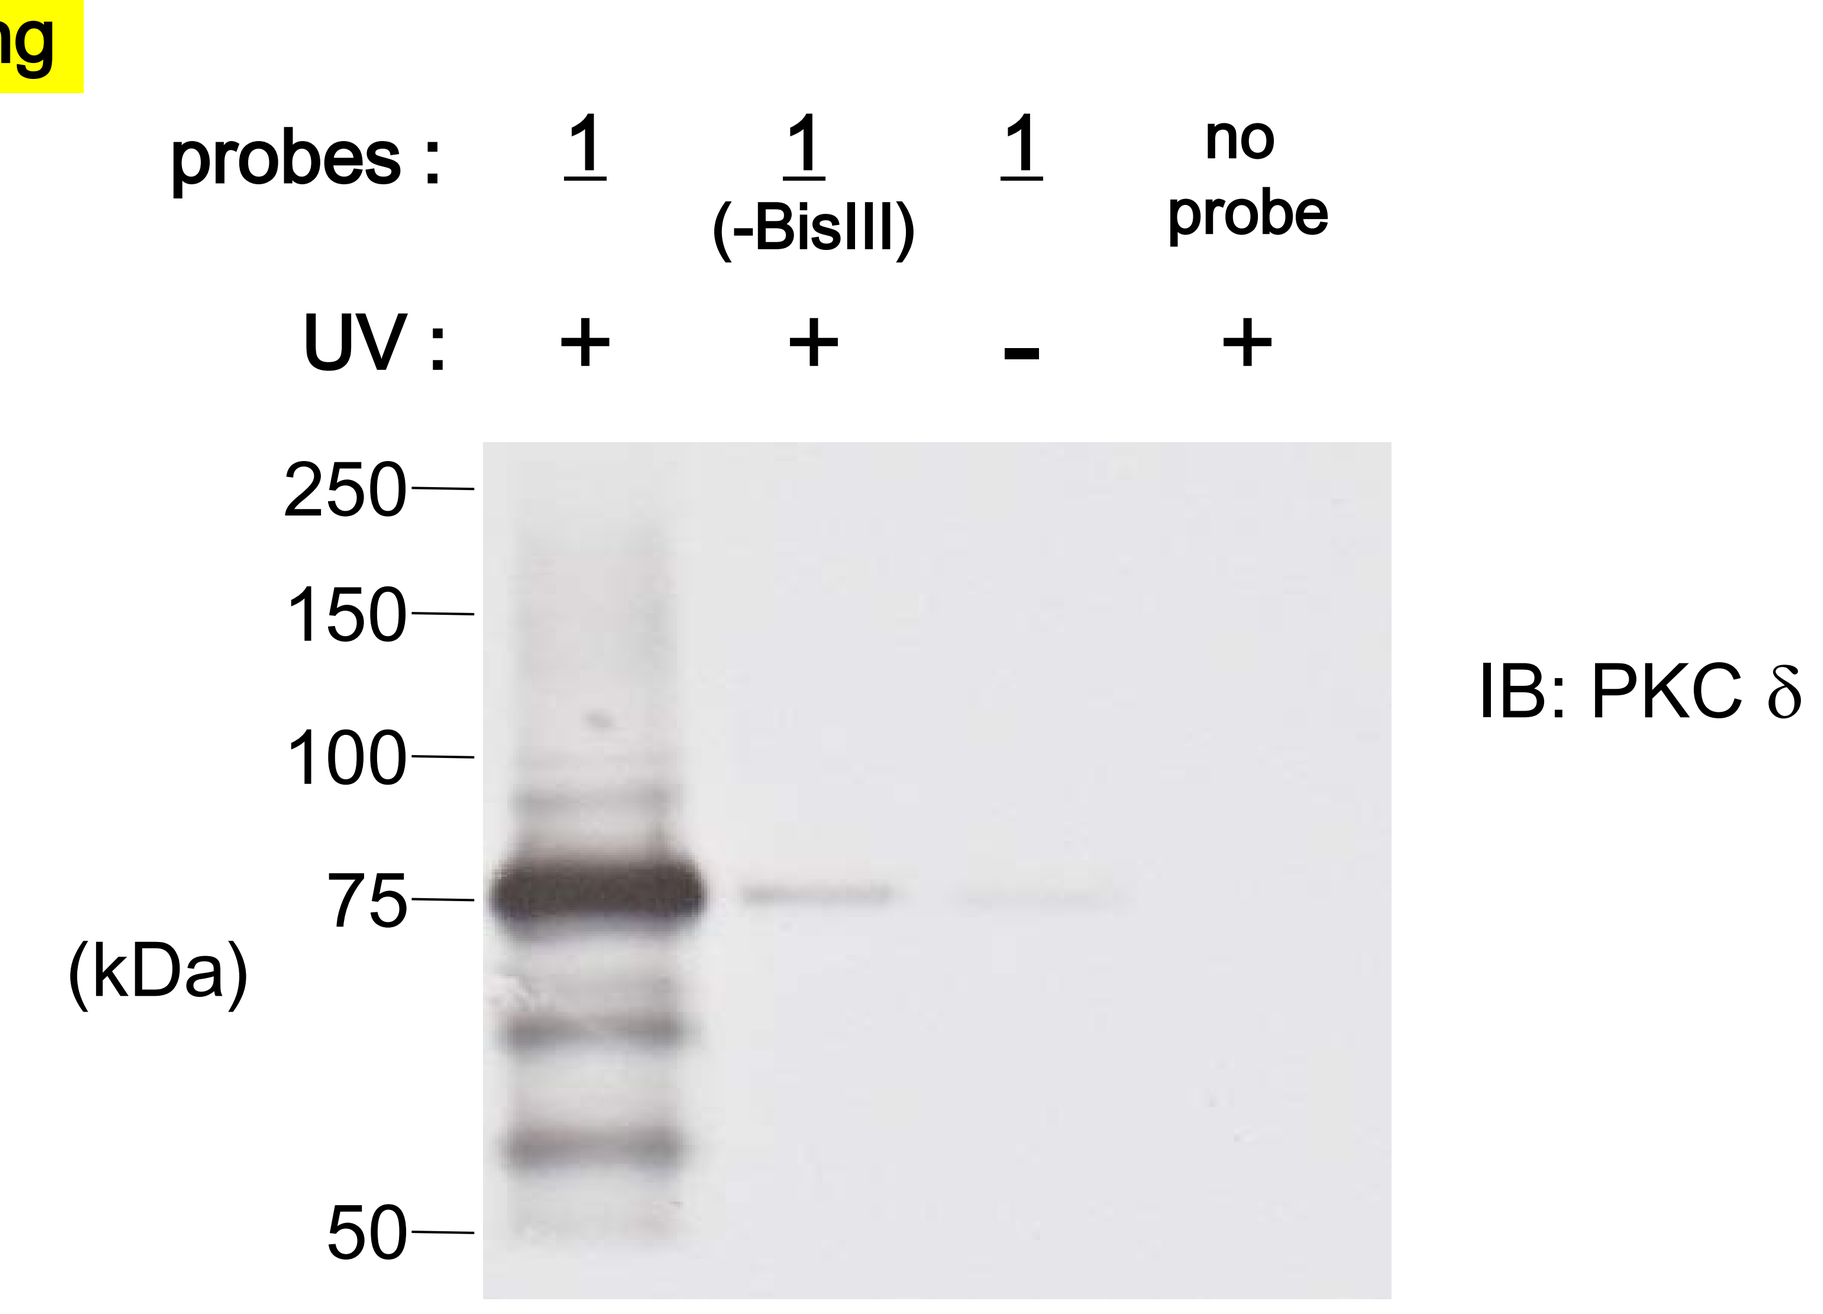
probes : 1 | 1 (-BisIII) | 1 | no probe
UV : + | + | - | +
250
150
100
75
50
(kDa)
IB: PKC δ

MS analysis (spectral counts)

	1 (UV +)	1 (-BisIII) (UV +)	1 (UV -)	no probe (UV +)	
PKCδ	28	0	0	0	target proteins
GSK3β	10	0	0	0	
PRKDC	75	25	0	4	non-specific proteins
P4HB	51	17	1	1	

effects of the competitor

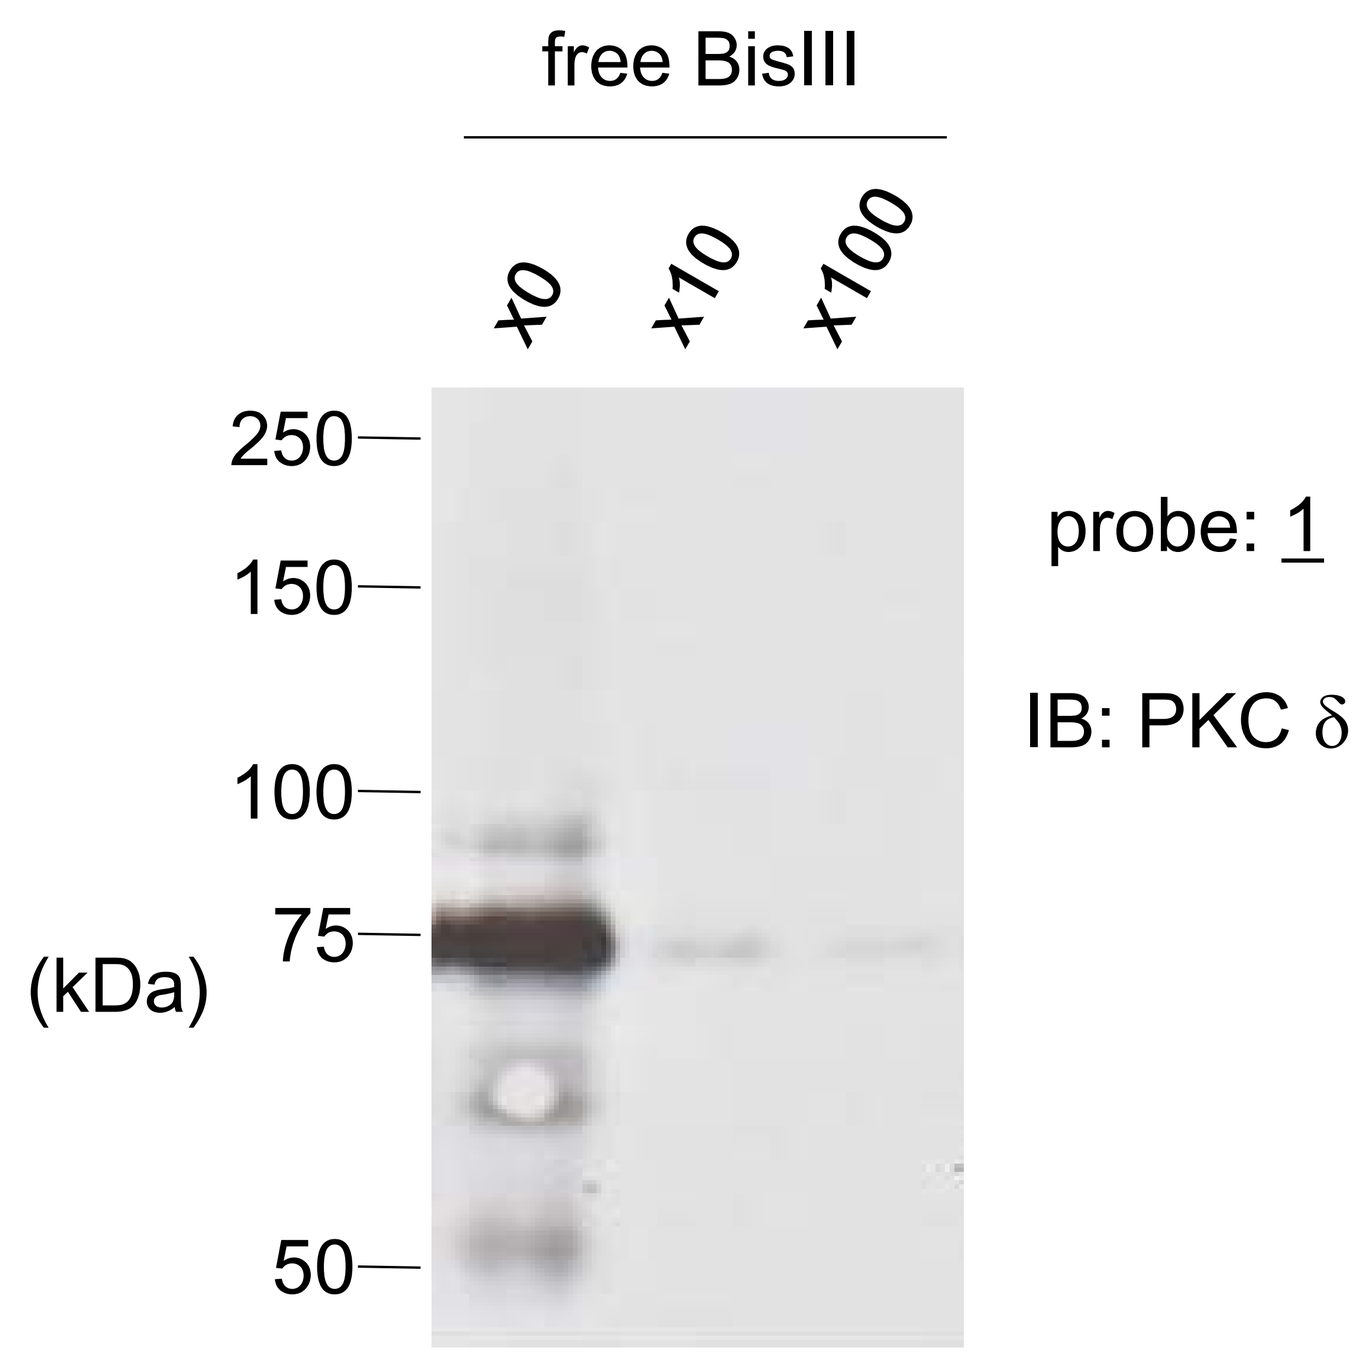
free BisIII
x0
x10
x100
250
150
100
75
50
(kDa)
probe: 1
IB: PKC δ

	x0	x10	x100	
PKCδ	23	0	0	target proteins
GSK3β	7	0	0	
PRKDC	14	29	17	non-specific proteins
P4HB	55	78	91	

Cross-linking was very effective to capture the BisIII target proteins.
The target proteins could be distinguished from non-specific ones by the competition experiments.

3. The linker property influenced the quantity of the captured proteins.

effects of the linker length (number of Gly units)

Probe No.	1	2	3	4
number of Gly units	0	1	2	3
linker length*	5	8	11	14

* length from the BisIII connection point to the main chain part in each probe

MS analysis (spectral counts)

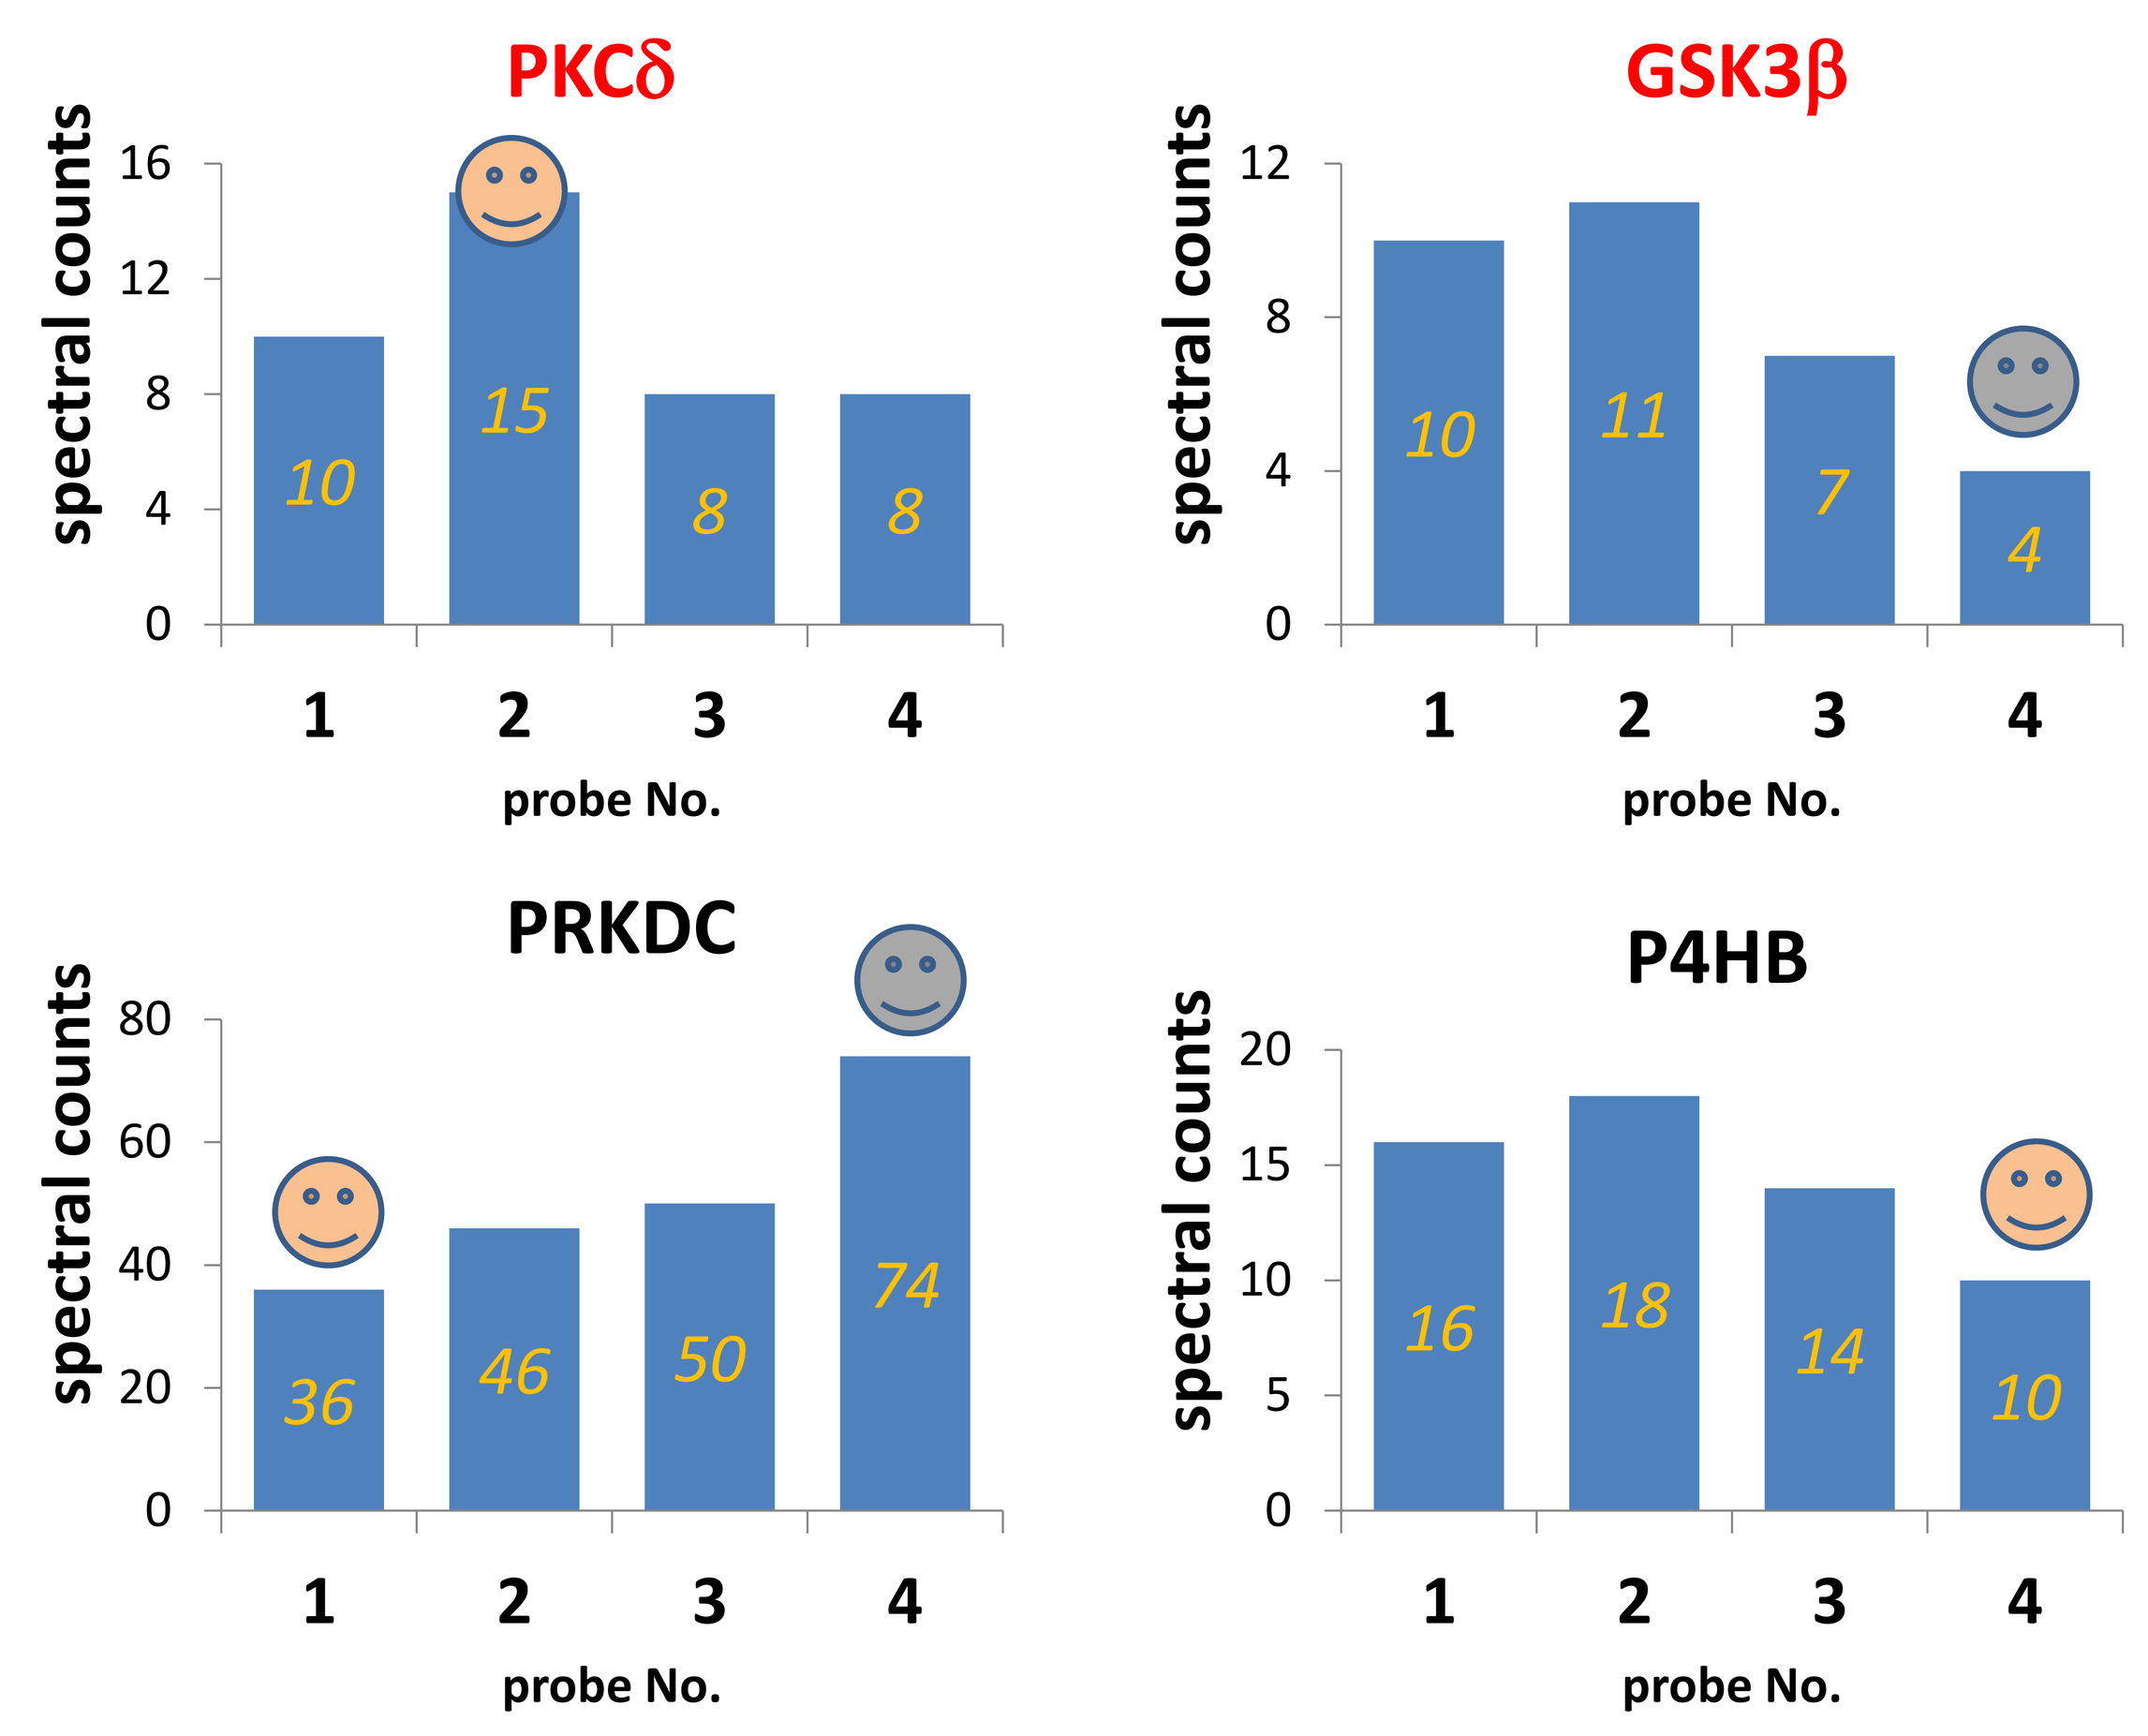
PKCδ
10
15
8
8
GSK3β
10
11
7
4
PRKDC
36
46
50
74
P4HB
16
18
14
10
spectral counts
probe No.

PKCδ and GSK3β showed different trends in this experiment.
PRKDC were increasing with the increasing of Gly units.

effects of physicochemical properties of the linkers

Probe No.	1	3	4	5	6	7
additional units	none	Gly x2	Gly x3	methylenes	ethylene glycol	amine+methylenes
linker length*	5	11	14	13	14	12

* length from the BisIII connection point to the main chain part in each probe

MS analysis (spectral counts)

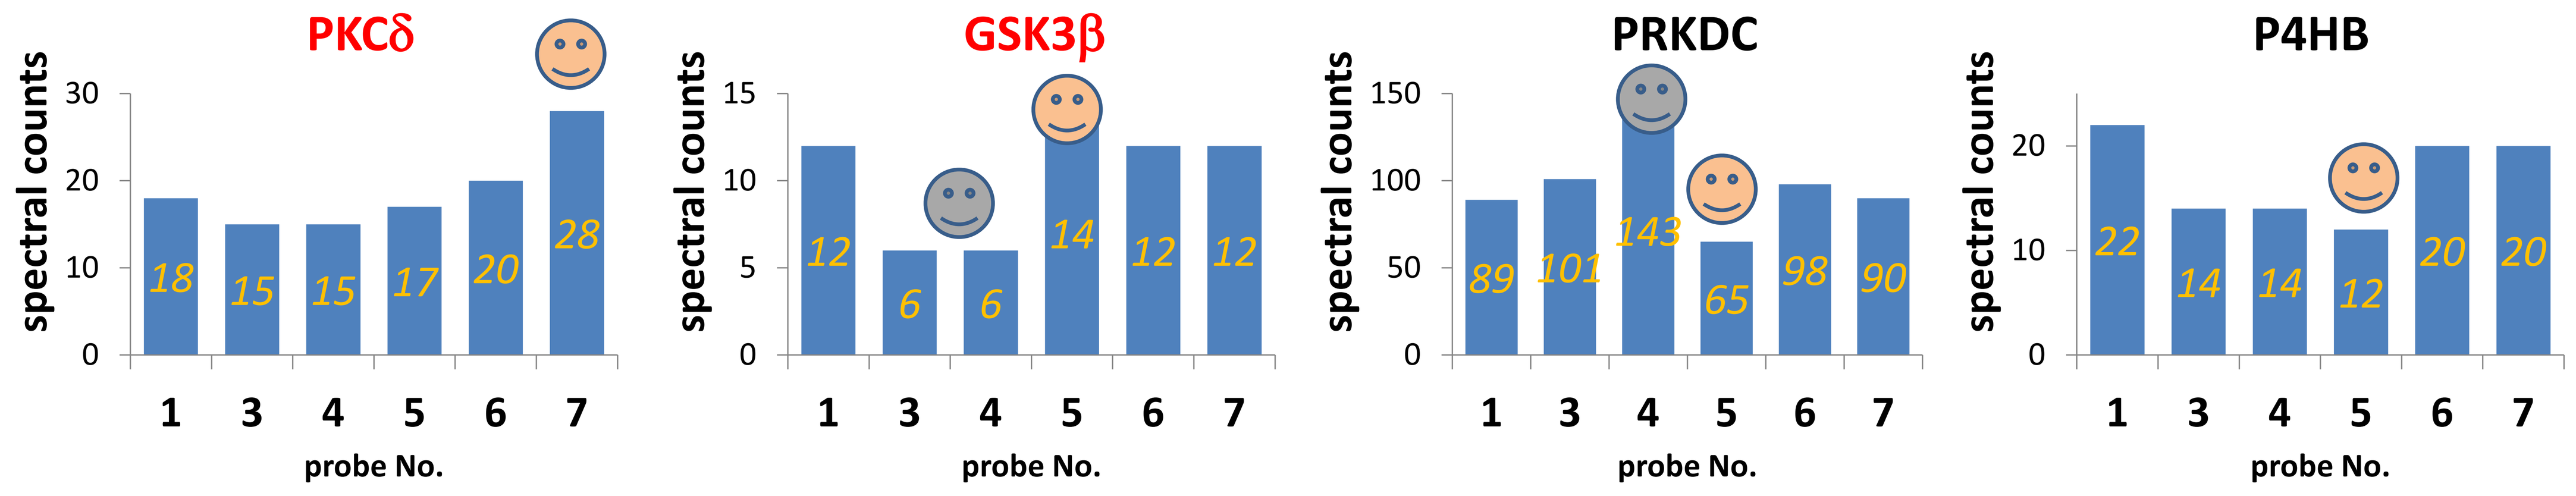
PKCδ
18
15
15
17
20
28
GSK3β
12
6
6
14
12
12
PRKDC
89
101
143
65
98
90
P4HB
22
14
14
12
20
20
spectral counts
probe No.

MS analysis (sum of peptides area)

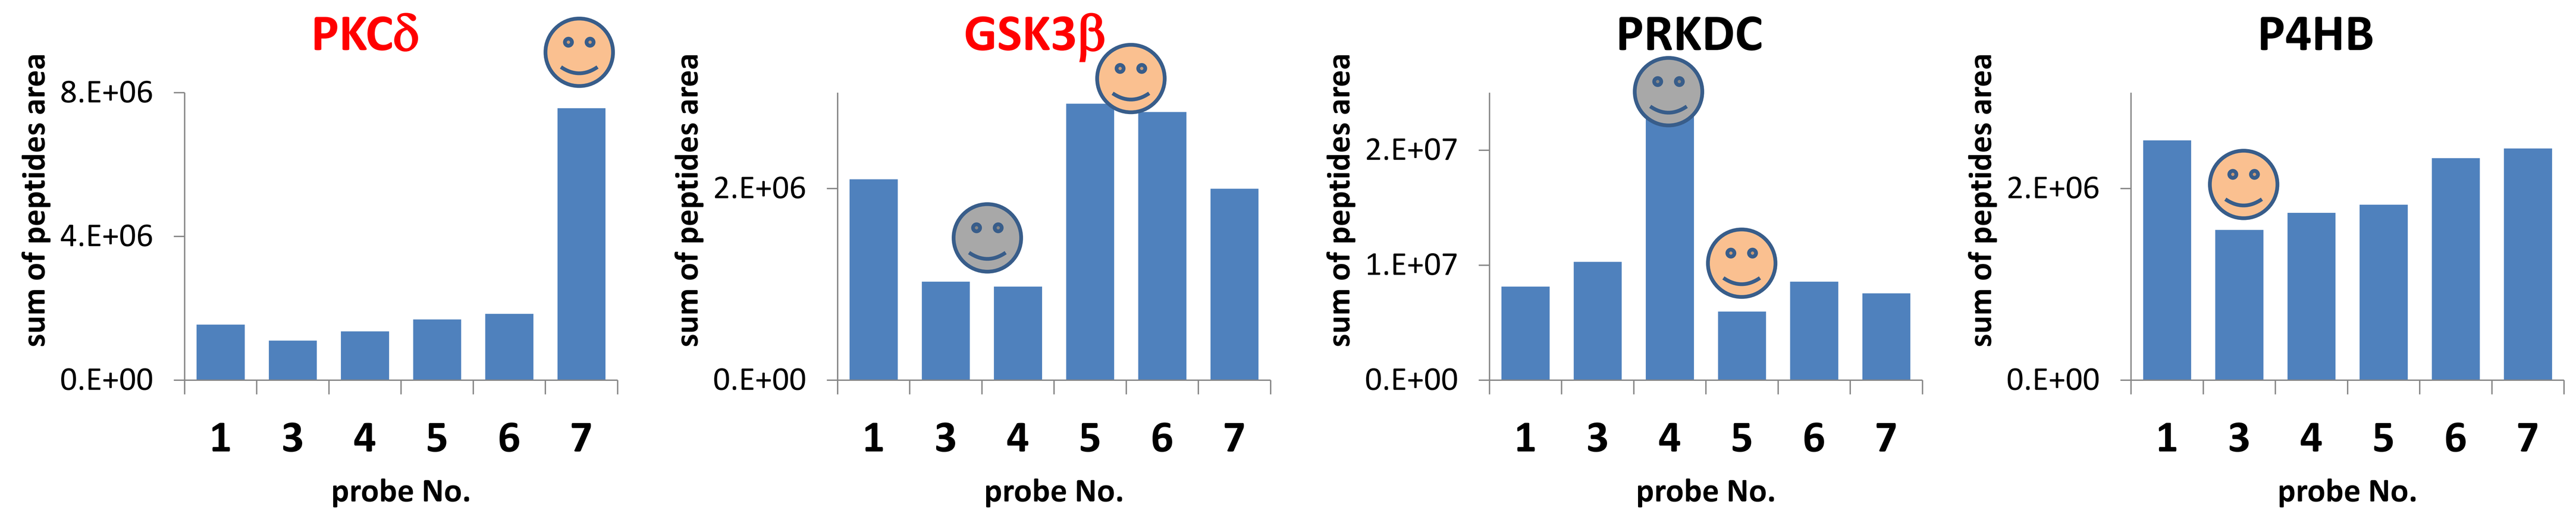
PKCδ
GSK3β
PRKDC
P4HB
sum of peptides area
probe No.

The flexible linker type probes 5, 6 and 7 showed good results for capturing the target proteins.
Probe 7 was the best for PKCδ, but was not for GSK3β.
There was a good correlation between the two MS analysis methods.

Methods

Pull down experiments with photoreaction

THP-1 cell lysates (500 μg in 0.2% CHAPS buffer) were pre-incubated with free BisIII or DMF for 30 min. After the probes (100 nmol) were added to the lysates, they were incubated for 2 hr at 4°C. Then the solutions were exposed to UV light (wave length 302 nm) for 1 min at 4°C. The solutions were incubated with Streptavidin beads for 30 min at 4°C, then the beads were collected and washed three times.

Protein digestion and Mass spectrometry

To the collected beads, digestion buffer and 0.5 μg of trypsin were added and they were incubated for 16 hr at 37°C. After dilution with acidic buffer, they were applied for the stage tip purification. The desalted samples were dissolved and loaded directly onto a C18 analytical column. LC/MS/MS experiments were performed on Ultimate 3000 liquid chromatography system (Dionex) connected with an LTQ Orbitrap XL mass spectrometer (Thermo Fisher) utilizing a nano-electrospray ion source. LC separation was performed at a flow rate 400 nl/min with linear gradient of 2-45 % LC-B over 80 min. Mass spectrometric analysis was performed on top3 data-dependent scan mode with the lock mass option.

Protein identification and label-free quantitative analysis

Protein identification was performed using Mascot software (Matrix Science) against human protein database based on NCBI REFSEQ protein sequence. Acetylation (protein N-term), oxidation (Met) and pyro-Glu (N-term Glu and Gln) were set for variable modifications. Precursor ion and fragment ion mass tolerances were set to 10 ppm and 0.8 Da, respectively. Up to three missed trypsin cleavage was allowed. Peptide identification criteria was $P<0.05$.

The data of spectral counts were summarized using in-house original program.

The data of sum of peptides area were calculated using the Progenesis LC-MS software (Nonlinear Dynamics).